63rd ERA CONGRESS
GLASGOW & VIRTUAL
JUNE 3-6, 2026
Challenge Your Thinking

Interim Results of a Phase 1 Study to Evaluate the Safety, Tolerability, Pharmacokinetics, and Pharmacodynamics of JADE101 in Healthy Volunteers.

B. GUFFORD[1], M. SOTO[1], C. PISCIA-NICHOLS[1], J. LEISKE[1], T. DEVRIES[1], V. TONG[1], S. LEWIS[1], L. LI[1], E. FILBERT[1], R. KATIAL[2], and A. KING[1]

[1] Jade Biosciences, San Francisco, United States of America and Vancouver, Canada, [2] New Zealand Clinical Research, Auckland, New Zealand

BACKGROUND AND AIMS

- A proliferation-inducing ligand (APRIL) plays a critical role in driving the production of pathogenic Gd-IgA1 in IgAN.
- JADE101 is a novel APRIL-neutralizing monoclonal antibody (mAb) designed with ultra-high affinity and extended half-life.
- The JADE101-01 First-In-Human (FIH) study aims to characterize the safety, tolerability, pharmacokinetics (PK), pharmacodynamics (PD), and immunogenicity of JADE101 following single subcutaneous (SC) doses.

JADE101 Delivers Compelling Magnitude and Duration of IgA Reduction with Modest Impact on IgG Following a Single Dose

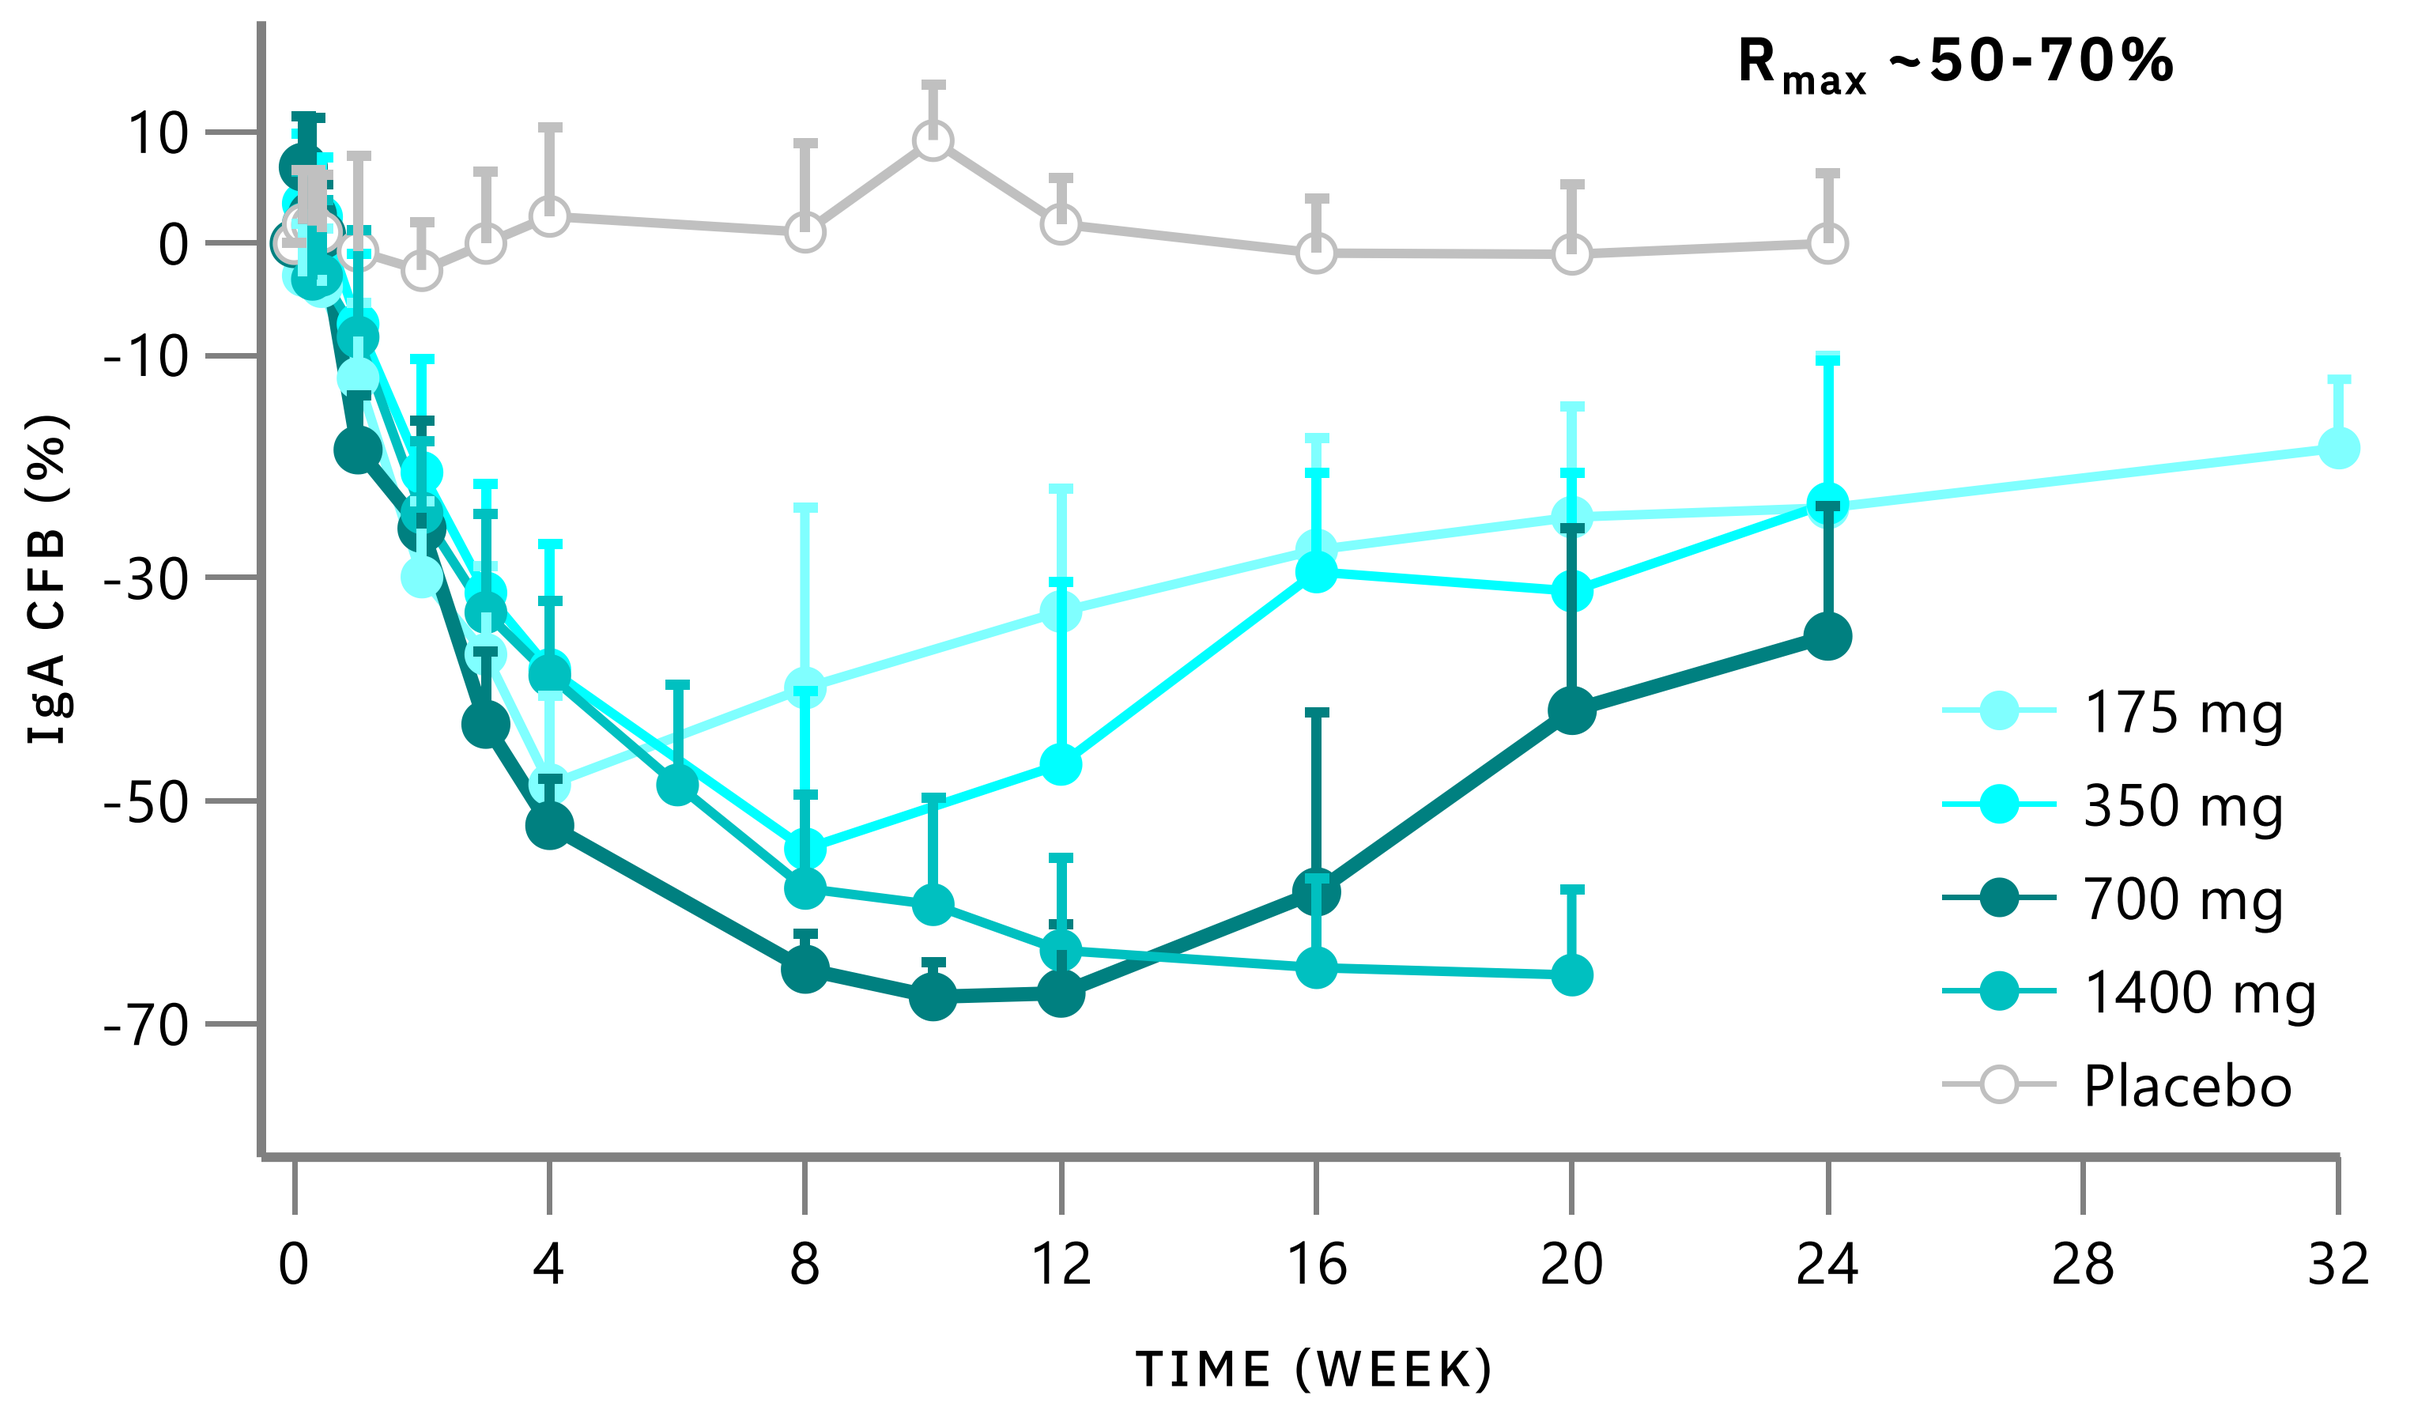
IgA
R_{max} ~50-70%
IgA CFB (%)
TIME (WEEK)
175 mg
350 mg
700 mg
1400 mg
Placebo

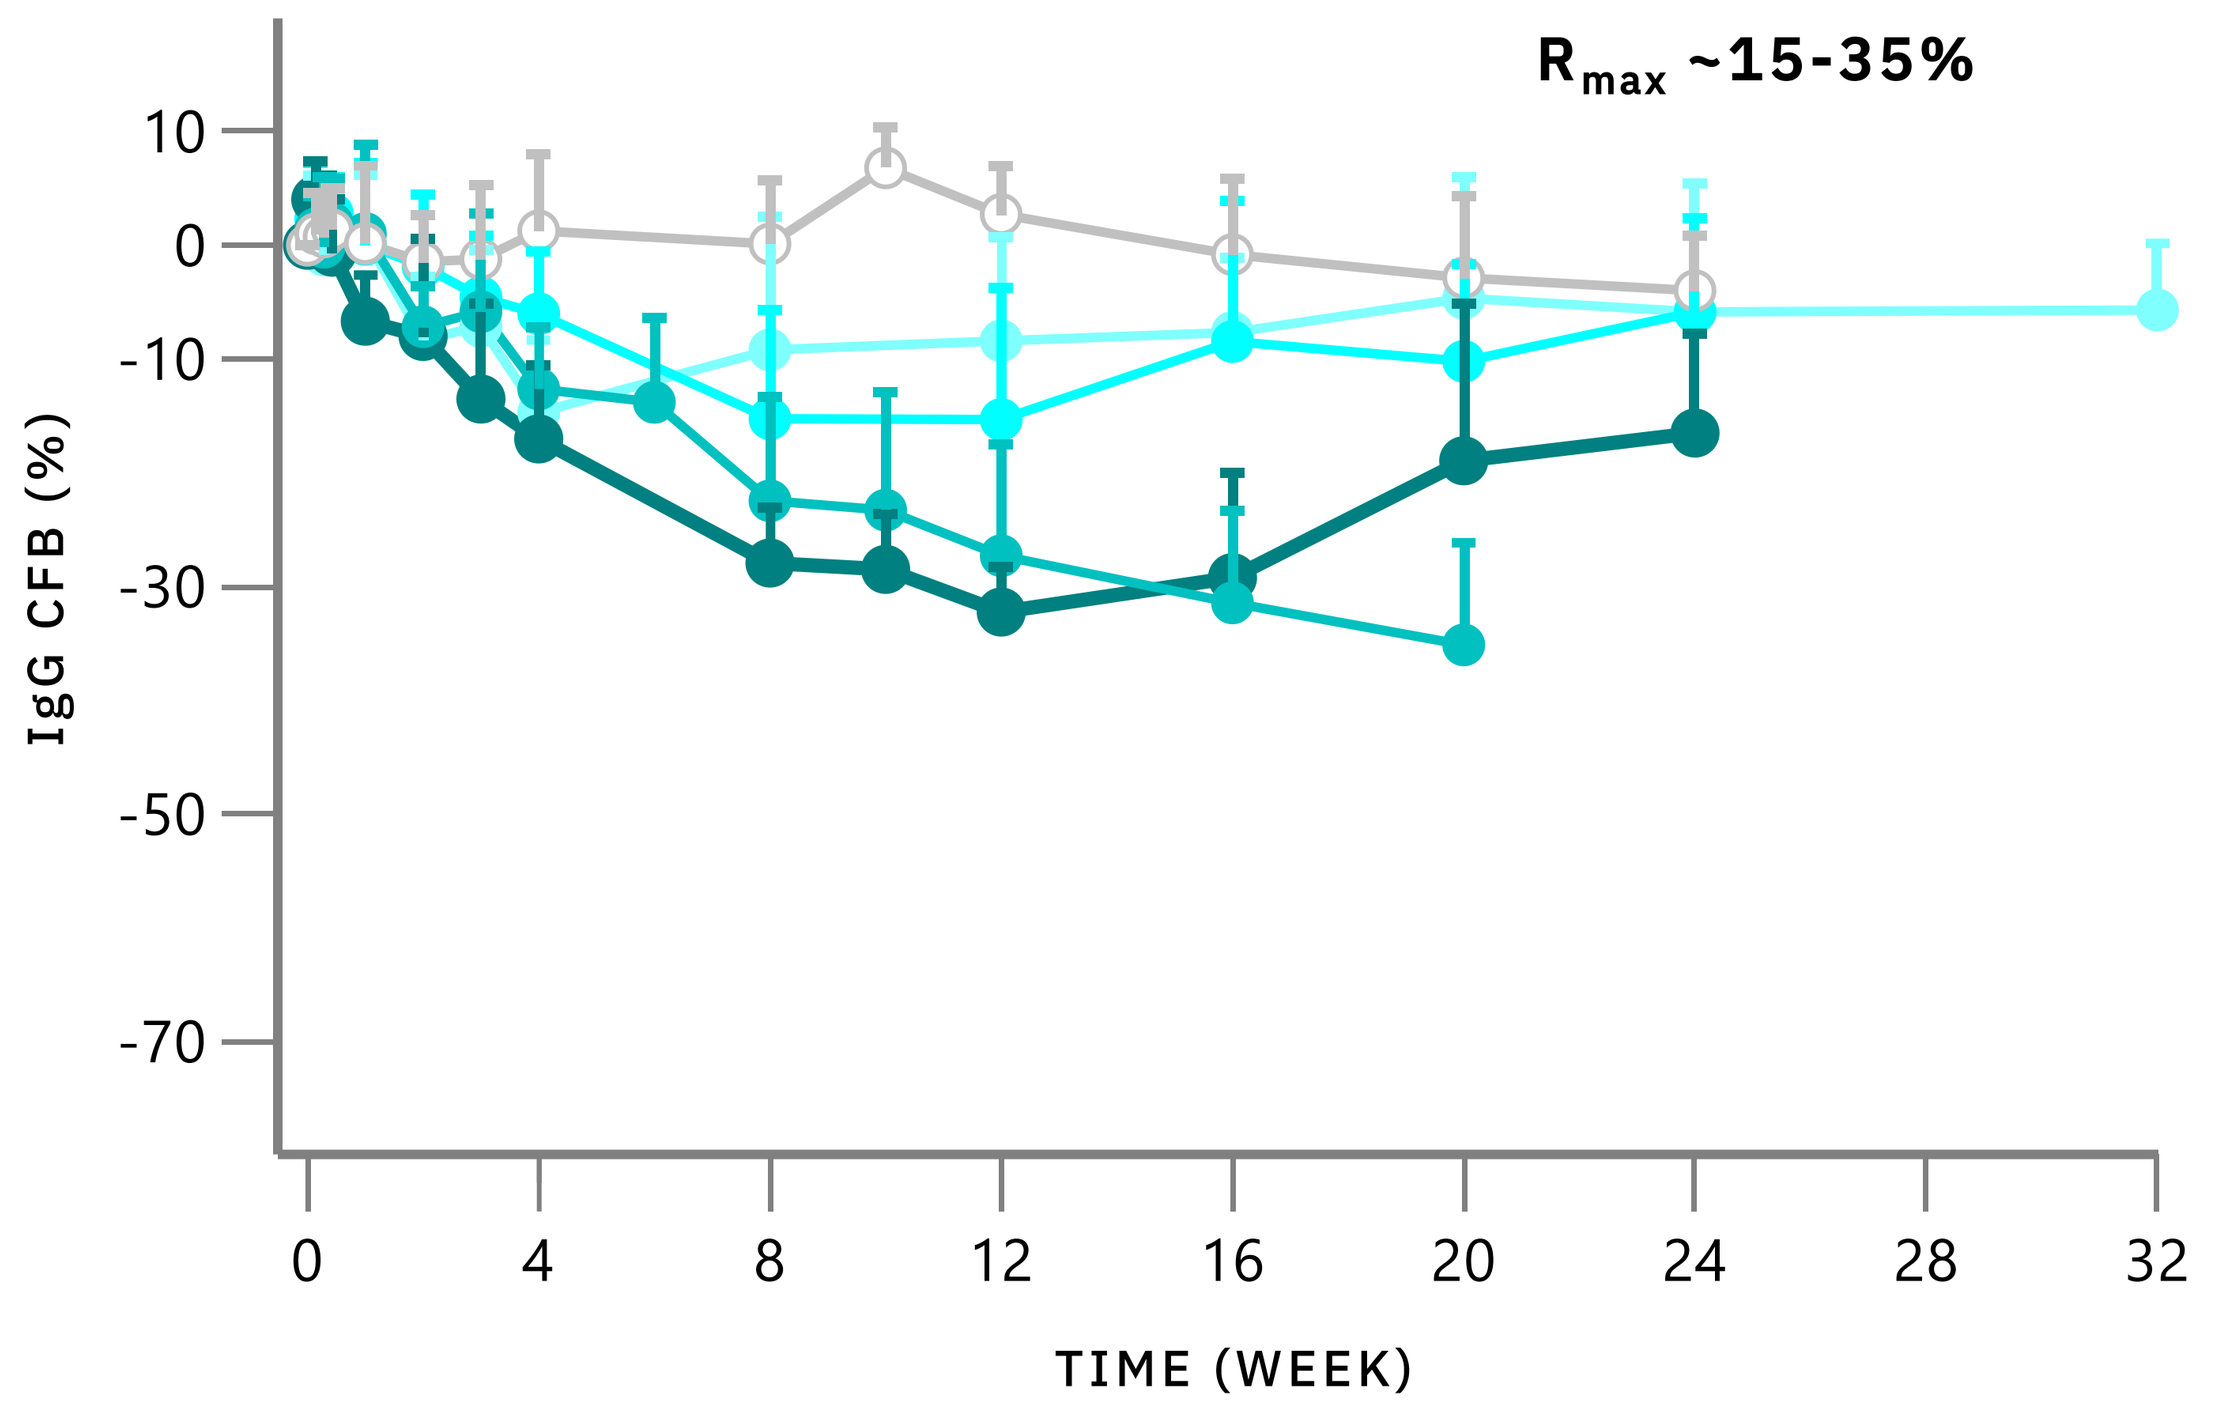
IgG
R_{max} ~15-35%
IgG CFB (%)
TIME (WEEK)

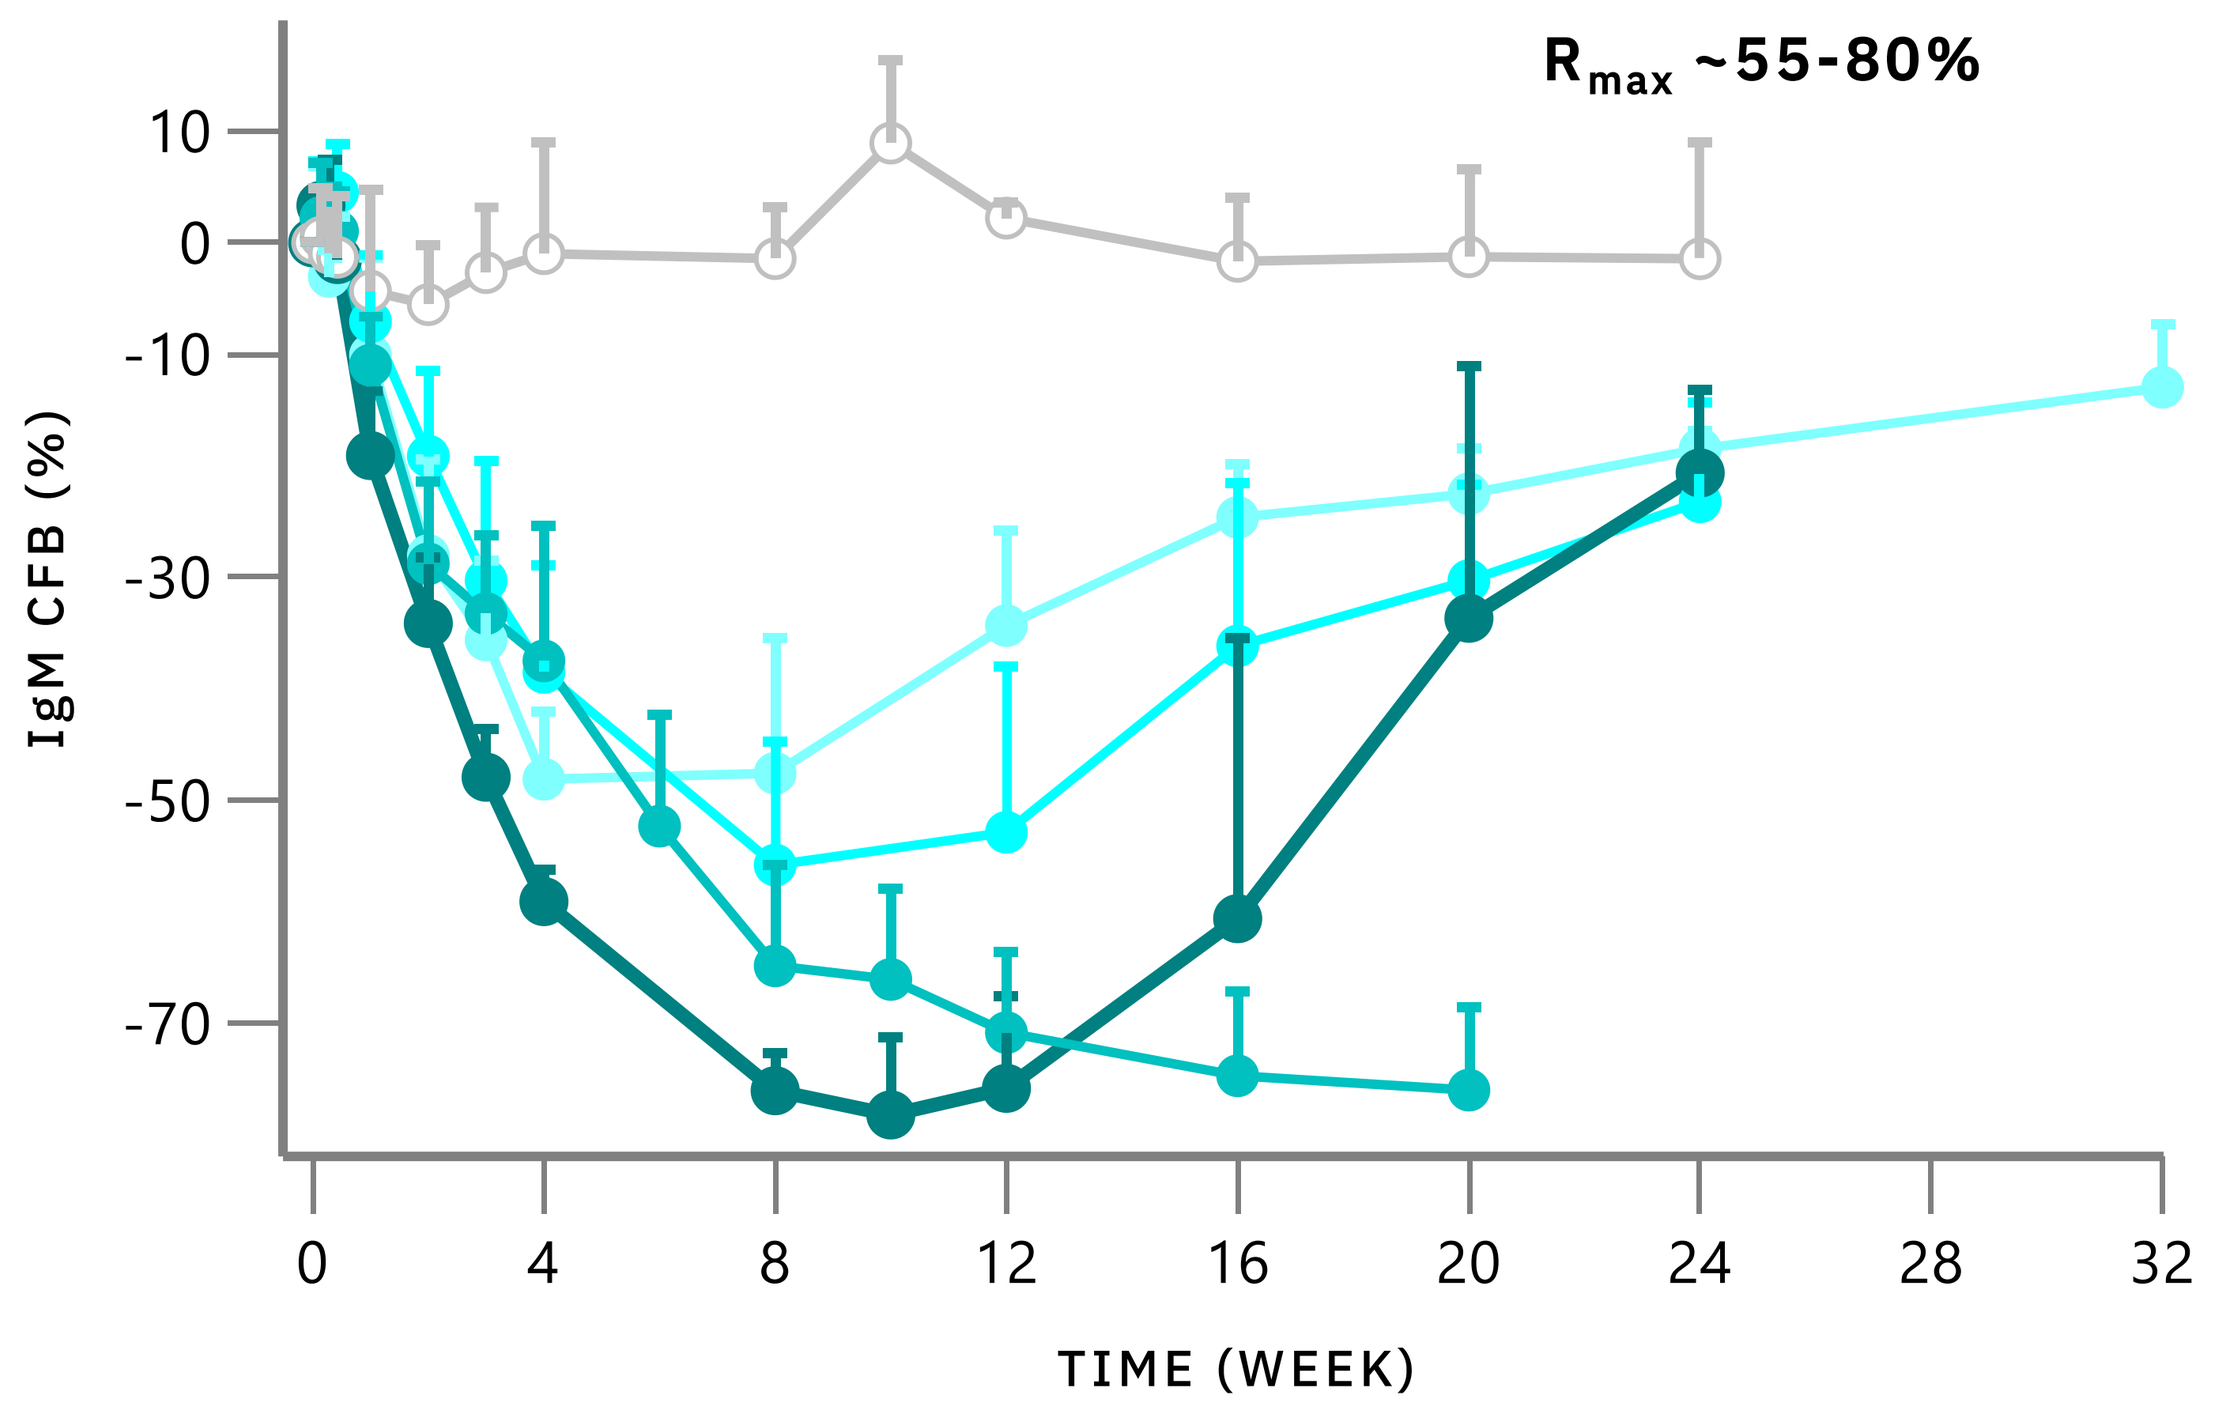
IgM
R_{max} ~55-80%
IgM CFB (%)
TIME (WEEK)

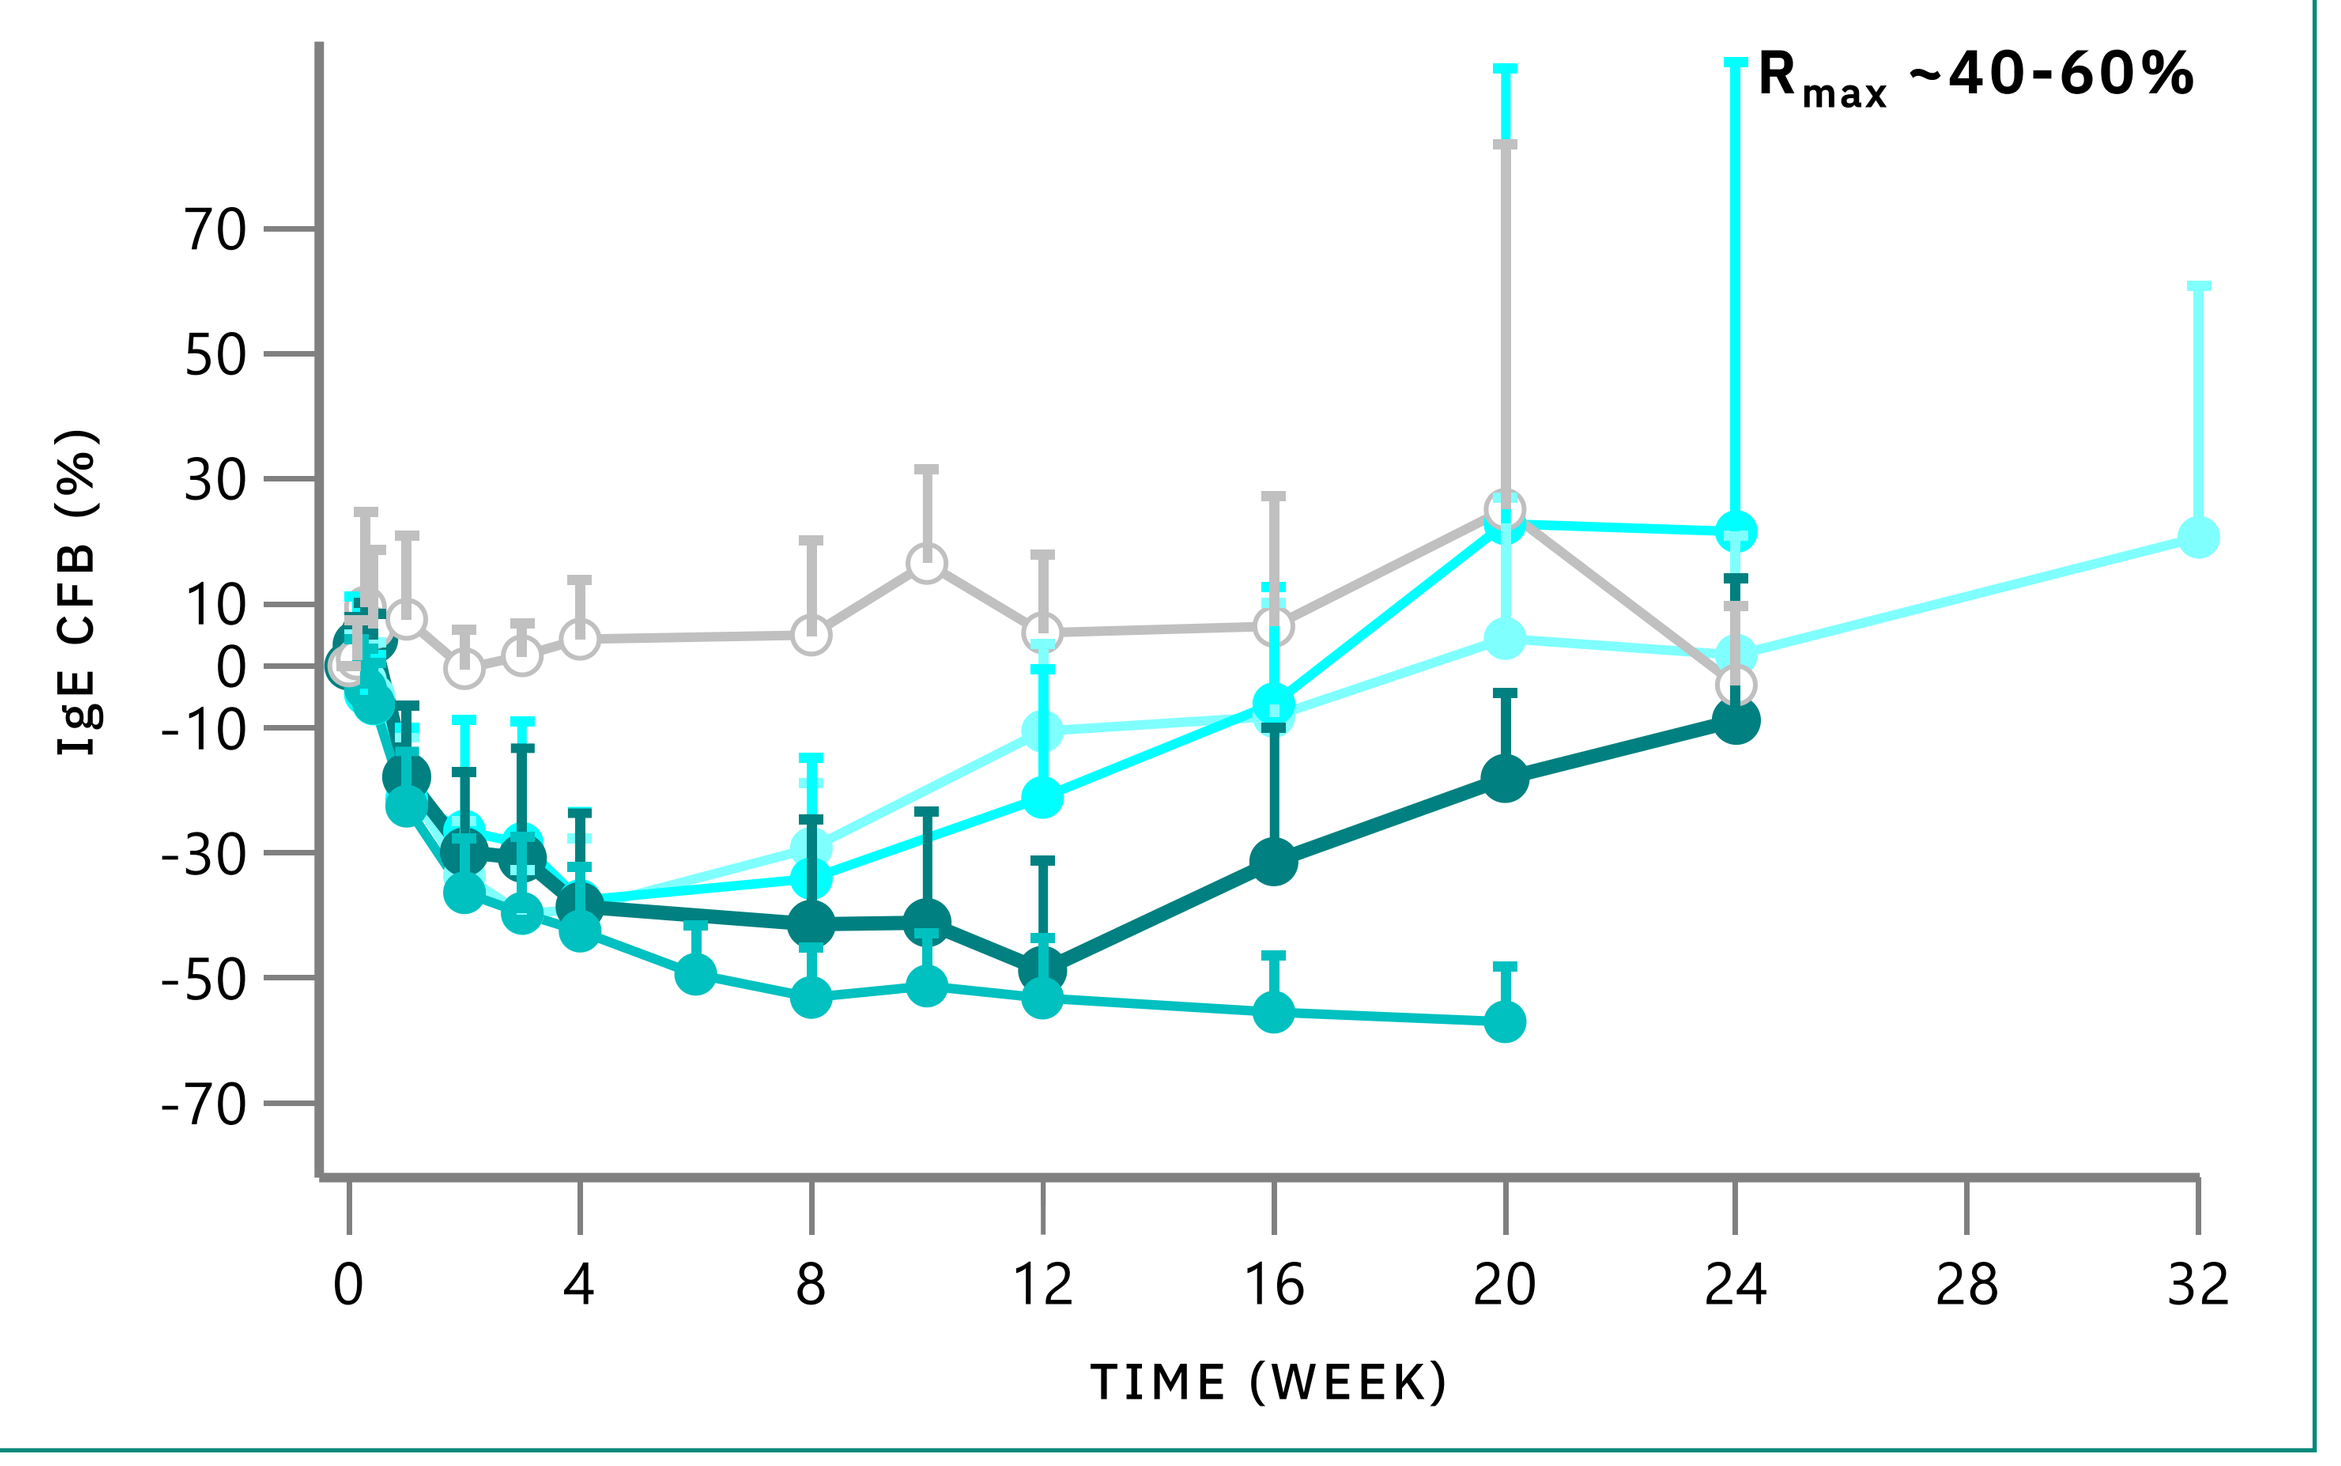
IgE
R_{max} ~40-60%
IgE CFB (%)
TIME (WEEK)

METHODS

- JADE101-01 is a Phase 1, randomized, double-blind, placebo-controlled, single-ascending dose study conducted at a single clinical research unit.

Study Population

Healthy Adult Volunteers (HVs)
N=8 per Cohort (6:2 active:placebo)

Endpoints

Primary: Safety and Tolerability

Secondary: Pharmacokinetics

Exploratory Pharmacodynamics: IgA, IgG, IgM, IgE, fAPRIL & Immunogenecity

Dose Levels and Interim Follow-up*

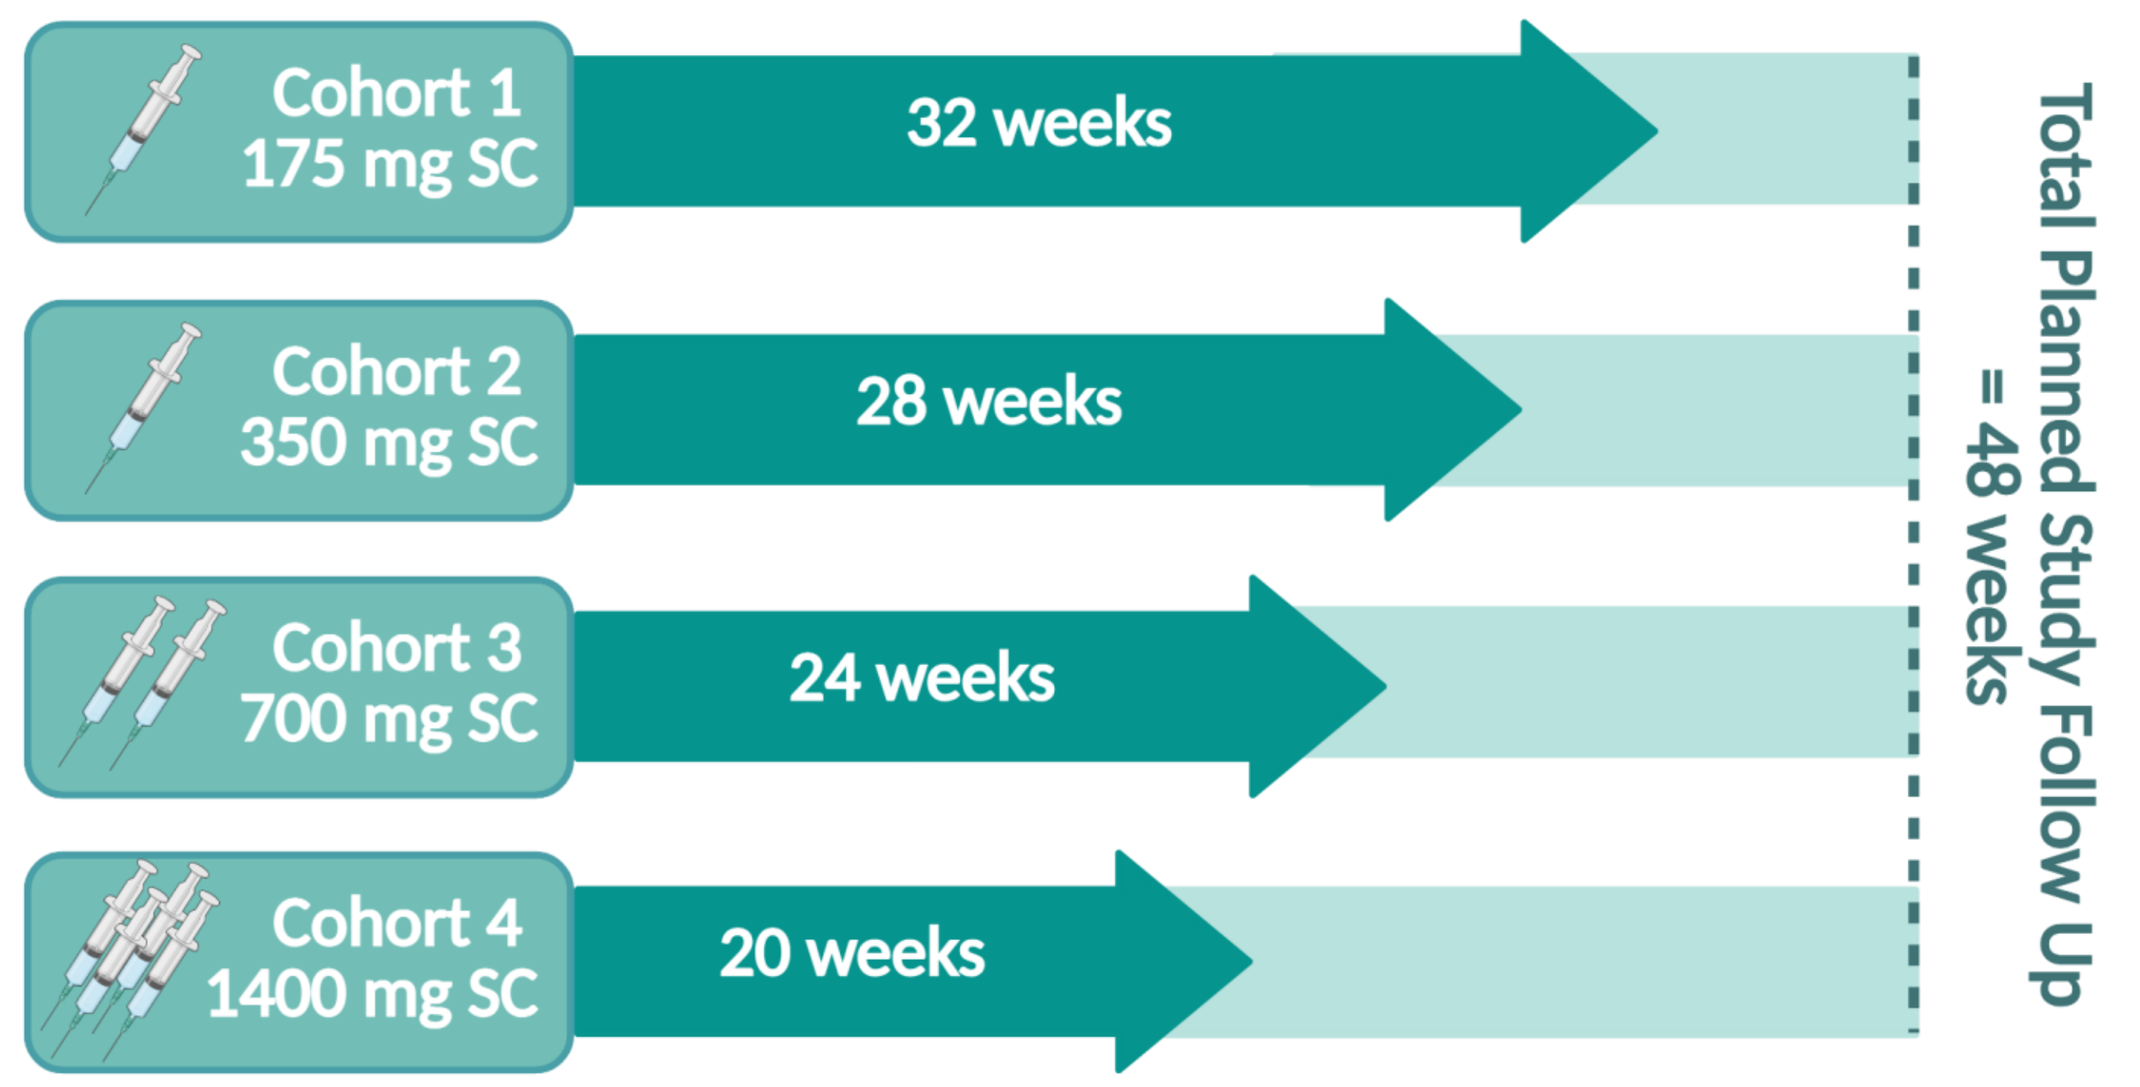
Cohort 1
175 mg SC
32 weeks
Cohort 2
350 mg SC
28 weeks
Cohort 3
700 mg SC
24 weeks
Cohort 4
1400 mg SC
20 weeks
Total Planned Study Follow Up
= 48 weeks

*Interim Data cut off: April 14, 2026

JADE101-01: NCT07059312

JADE101 Exhibits Dose-Dependent Extended PK Exposure, Rapid and Potent fAPRIL Neutralization

Pharmacokinetics

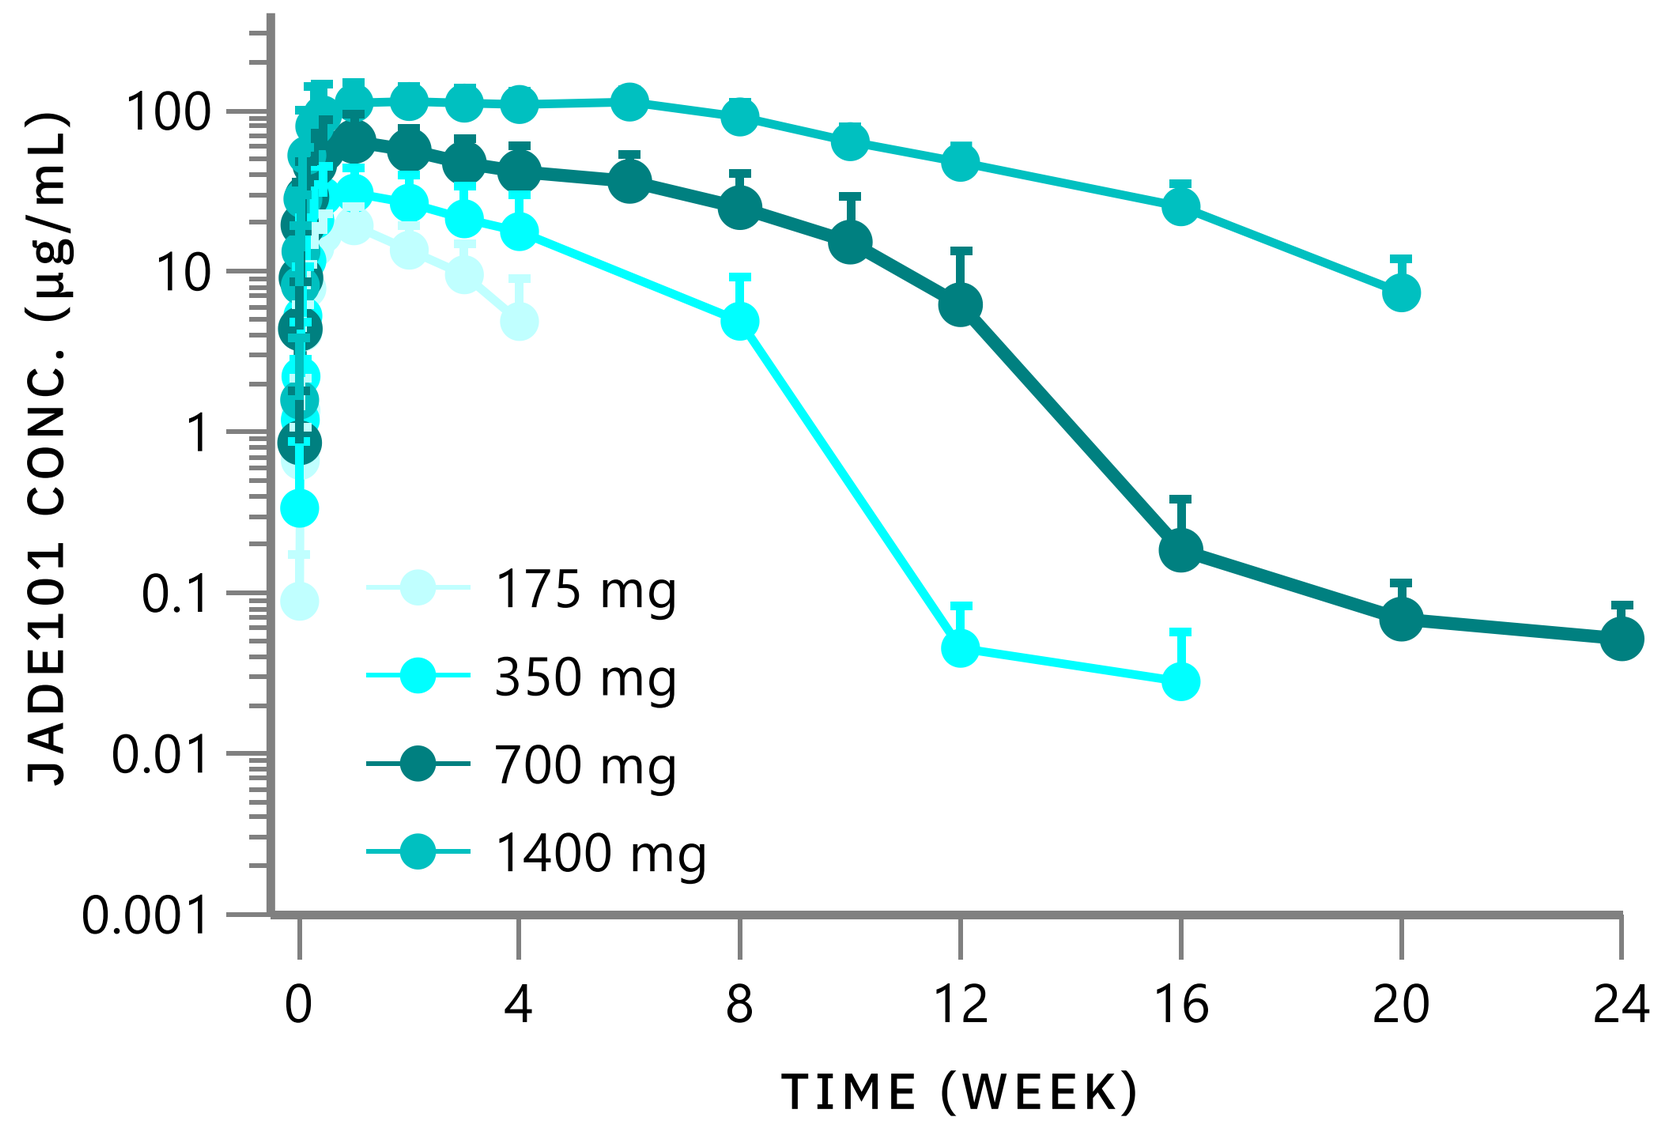
JADE101 CONC. (µg/mL)
TIME (WEEK)
175 mg
350 mg
700 mg
1400 mg

JADE101 Dose Level (N=6)	Parameter (units) C_{max} (µg/mL)	T_{max} (day)	AUC_{0-12W} (day·µg/mL)
175 mg	18.6 (33.9)	5.00 (3.00-7.00)	427 (44.6)
350 mg	27.8 (66.2)	4.96 (3.00-6.99)	761 (109.7)
700 mg	61.0 (49.7)	7.03 (3.00-21.0)	2540 (67.9)
1400 mg	120 (26.4)	24.4 (3.00-41.0)	7540 (26.0)

PK parameters presented as geometric mean (CV%) with the exception of T_{max} where median (min-max) presented. SC = subcutaneous

PK and PD data plotted as Mean + SD. IgA/G/M/E R_{max} range corresponds to mean maximum observed percent change from baseline across dose levels.

fAPRIL

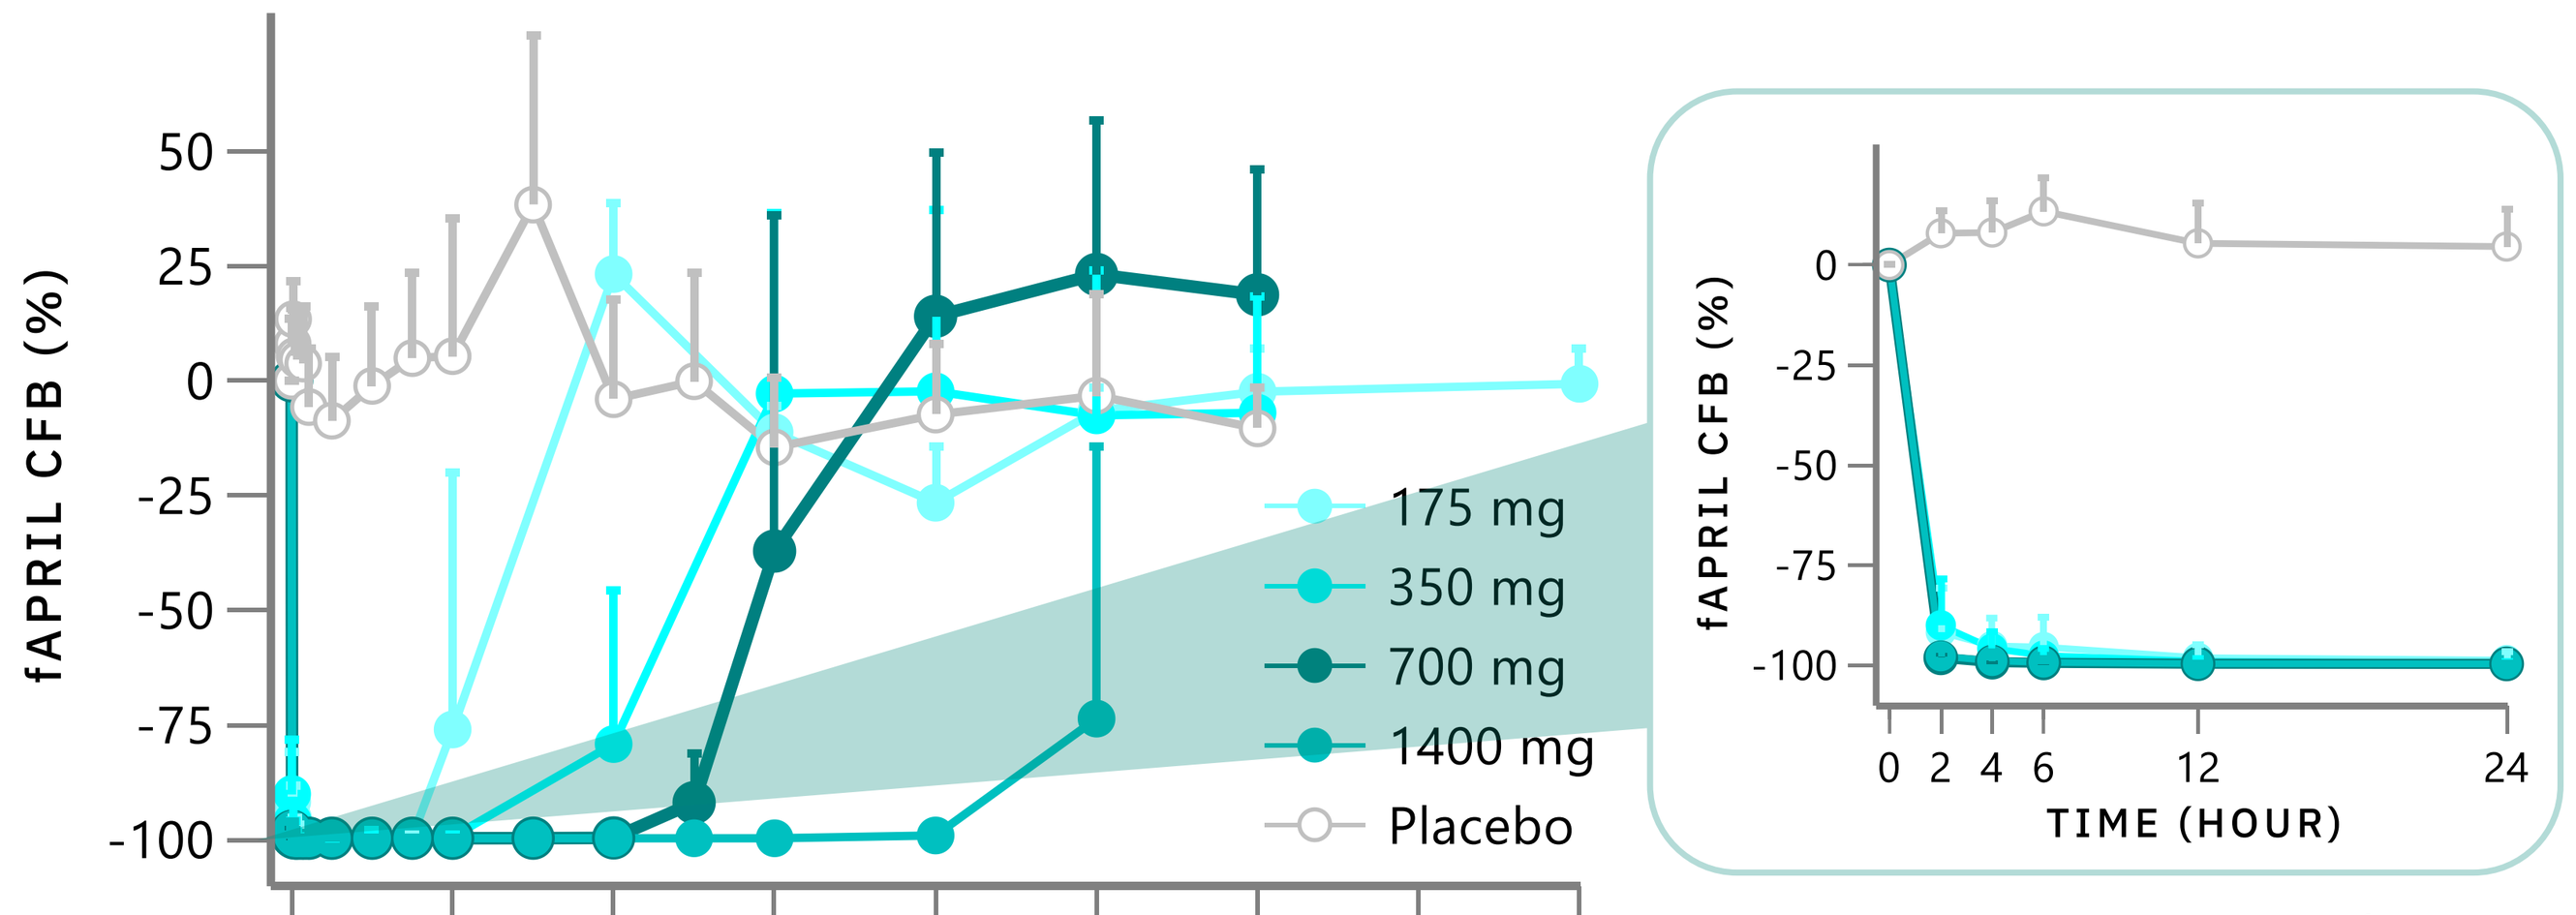
fAPRIL CFB (%)
TIME (WEEK)
175 mg
350 mg
700 mg
1400 mg
Placebo
TIME (HOUR)

Inset represents initial 24 hours following single SC dose administration

JADE101 is Generally Safe and Well-Tolerated at all Tested Doses in Healthy Volunteers

Demographics

JADE101 and placebo	175 mg (N = 8)	350 mg (N = 8)	700 mg (N = 8)	1400 mg (N = 8)	Overall (N = 32)
Age, (yr), Mean (SD)	38.0 (7.86)	44.5 (9.87)	28.0 (9.04)	37.6 (13.70)	37.0 (11.51)
Female N (%)	2 (25)	6 (75)	3 (38)	5 (63)	16 (50)
White N(%)	5 (63)	6 (75)	4 (50)	3 (38)	18 (56)
Asian N (%)	2 (25)	1 (13)	3 (38)	1 (13)	7 (22)
BMI, (kg/m²) Mean (SD)	25.5 (2.70)	26.7 (2.87)	25.9 (3.05)	24.2 (3.25)	25.6 (2.97)

Safety

JADE101 and placebo	175 mg (N = 8)	350 mg (N = 8)	700 mg (N = 8)	1400 mg (N = 8)	Overall (N = 32)
≥1 TEAE	6 (75.0%)	6 (75.0%)	5 (62.5%)	7 (87.5%)	24 (75.0%)
≥1 SAE	0%	0%	0%	0%	0%
≥1 severe TEAE	0%	0%	0%	0%	0%
Discontinued due to TEAE	0%	0%	0%	0%	0%
TEAEs occurring in ≥ 2 participants	Headache (8 participants; 25%); upper respiratory tract infection (7 participants; 21.9%); injection site erythema, oropharyngeal pain, and pyrexia (3 participants; 9.4%); abdominal pain upper, medical device site dermatitis, medical device site rash, musculoskeletal stiffness, nausea, and presyncope (2 participants each; 6.3%)				
Local tolerability / Immunogenicity	Well-tolerated locally by SC injection with mild (9%)/moderate(3%) injection site erythema, mild injection site pain (3%) and bruising (3%). / No clinically meaningful immunogenicity, and no apparent impact of ADA on PK/PD.				

CONCLUSIONS

- Interim results from the FIH study support a favorable safety and tolerability profile with rapid circulating fAPRIL suppression driving deep and prolonged IgA reduction after a single dose of JADE101.
- Biomarker responses in HVs is anticipated to translate to patients with IgAN, leading to potent suppression of IgA and Gd-IgA1 production, reduction in proteinuria and stabilization of eGFR.
- The biomarker-rich and translationally predictive data from this FIH evaluation establishes the foundation for subsequent clinical development of JADE101, a potentially disease-modifying therapy in IgAN with convenient, SC dosing being evaluated as infrequently as every 12 weeks.
- The Phase 2 evaluation of JADE101 in participants with IgAN is ongoing (NCT07541287).

ACKNOWLEDGEMENTS

Jade Biosciences thanks the healthy volunteer participants in JADE101-01 and our CRO/CRU partners at Fortrea/New Zealand Clinical Research.

QUESTIONS

EMAIL: INFO@JADEBIOSCIENCES.COM